Abstract # 3544768:

Post-Hoc Analysis of Suprachoroidal Injection Experience for Non-Infectious Uveitis

Milan Shah[1], Cherry Wan[2,F,E], Barry Kapik[2, F, E], Thomas A. Ciulla[2, F, E]

1.; Midwest Eye Institute, Indianapolis, IN 2. Clearside Biomedical, Alpharetta, Georgia

Disclosures: F (Financial Support); I (Personal Financial Interest); E (Employment); C (Consultant); P (Patent); R (Recipient)

E-mail: mLnshah@yahoo.com:

Purpose

- To analyze procedural characteristics of suprachoroidal injections (SCIs) using the SCS Microinjector® from data in two uveitis trials.
- The SCS Microinjector reliably delivers drug to the suprachoroidal space (SCS), an alternative administration route for chorioretinal diseases.
- SCIs were first attempted with a 900μm length needle and switched to 1100μm length if required. Correlations between needle length, baseline patient characteristics, and physician experience, via survey, with the device are presented.

Methods

- Post hoc analyses were performed to assess the relationship between needle length for baseline SCI and patient characteristics.
- Univariate analysis was conducted with Pearson chi-square analysis for categorial variables and the biserial correlation for continuous variables.
- Multivariate logistical regression was run to confirm univariate findings.
- Furthermore, a user experience survey was completed to evaluate real-world SCI experience in one trial.

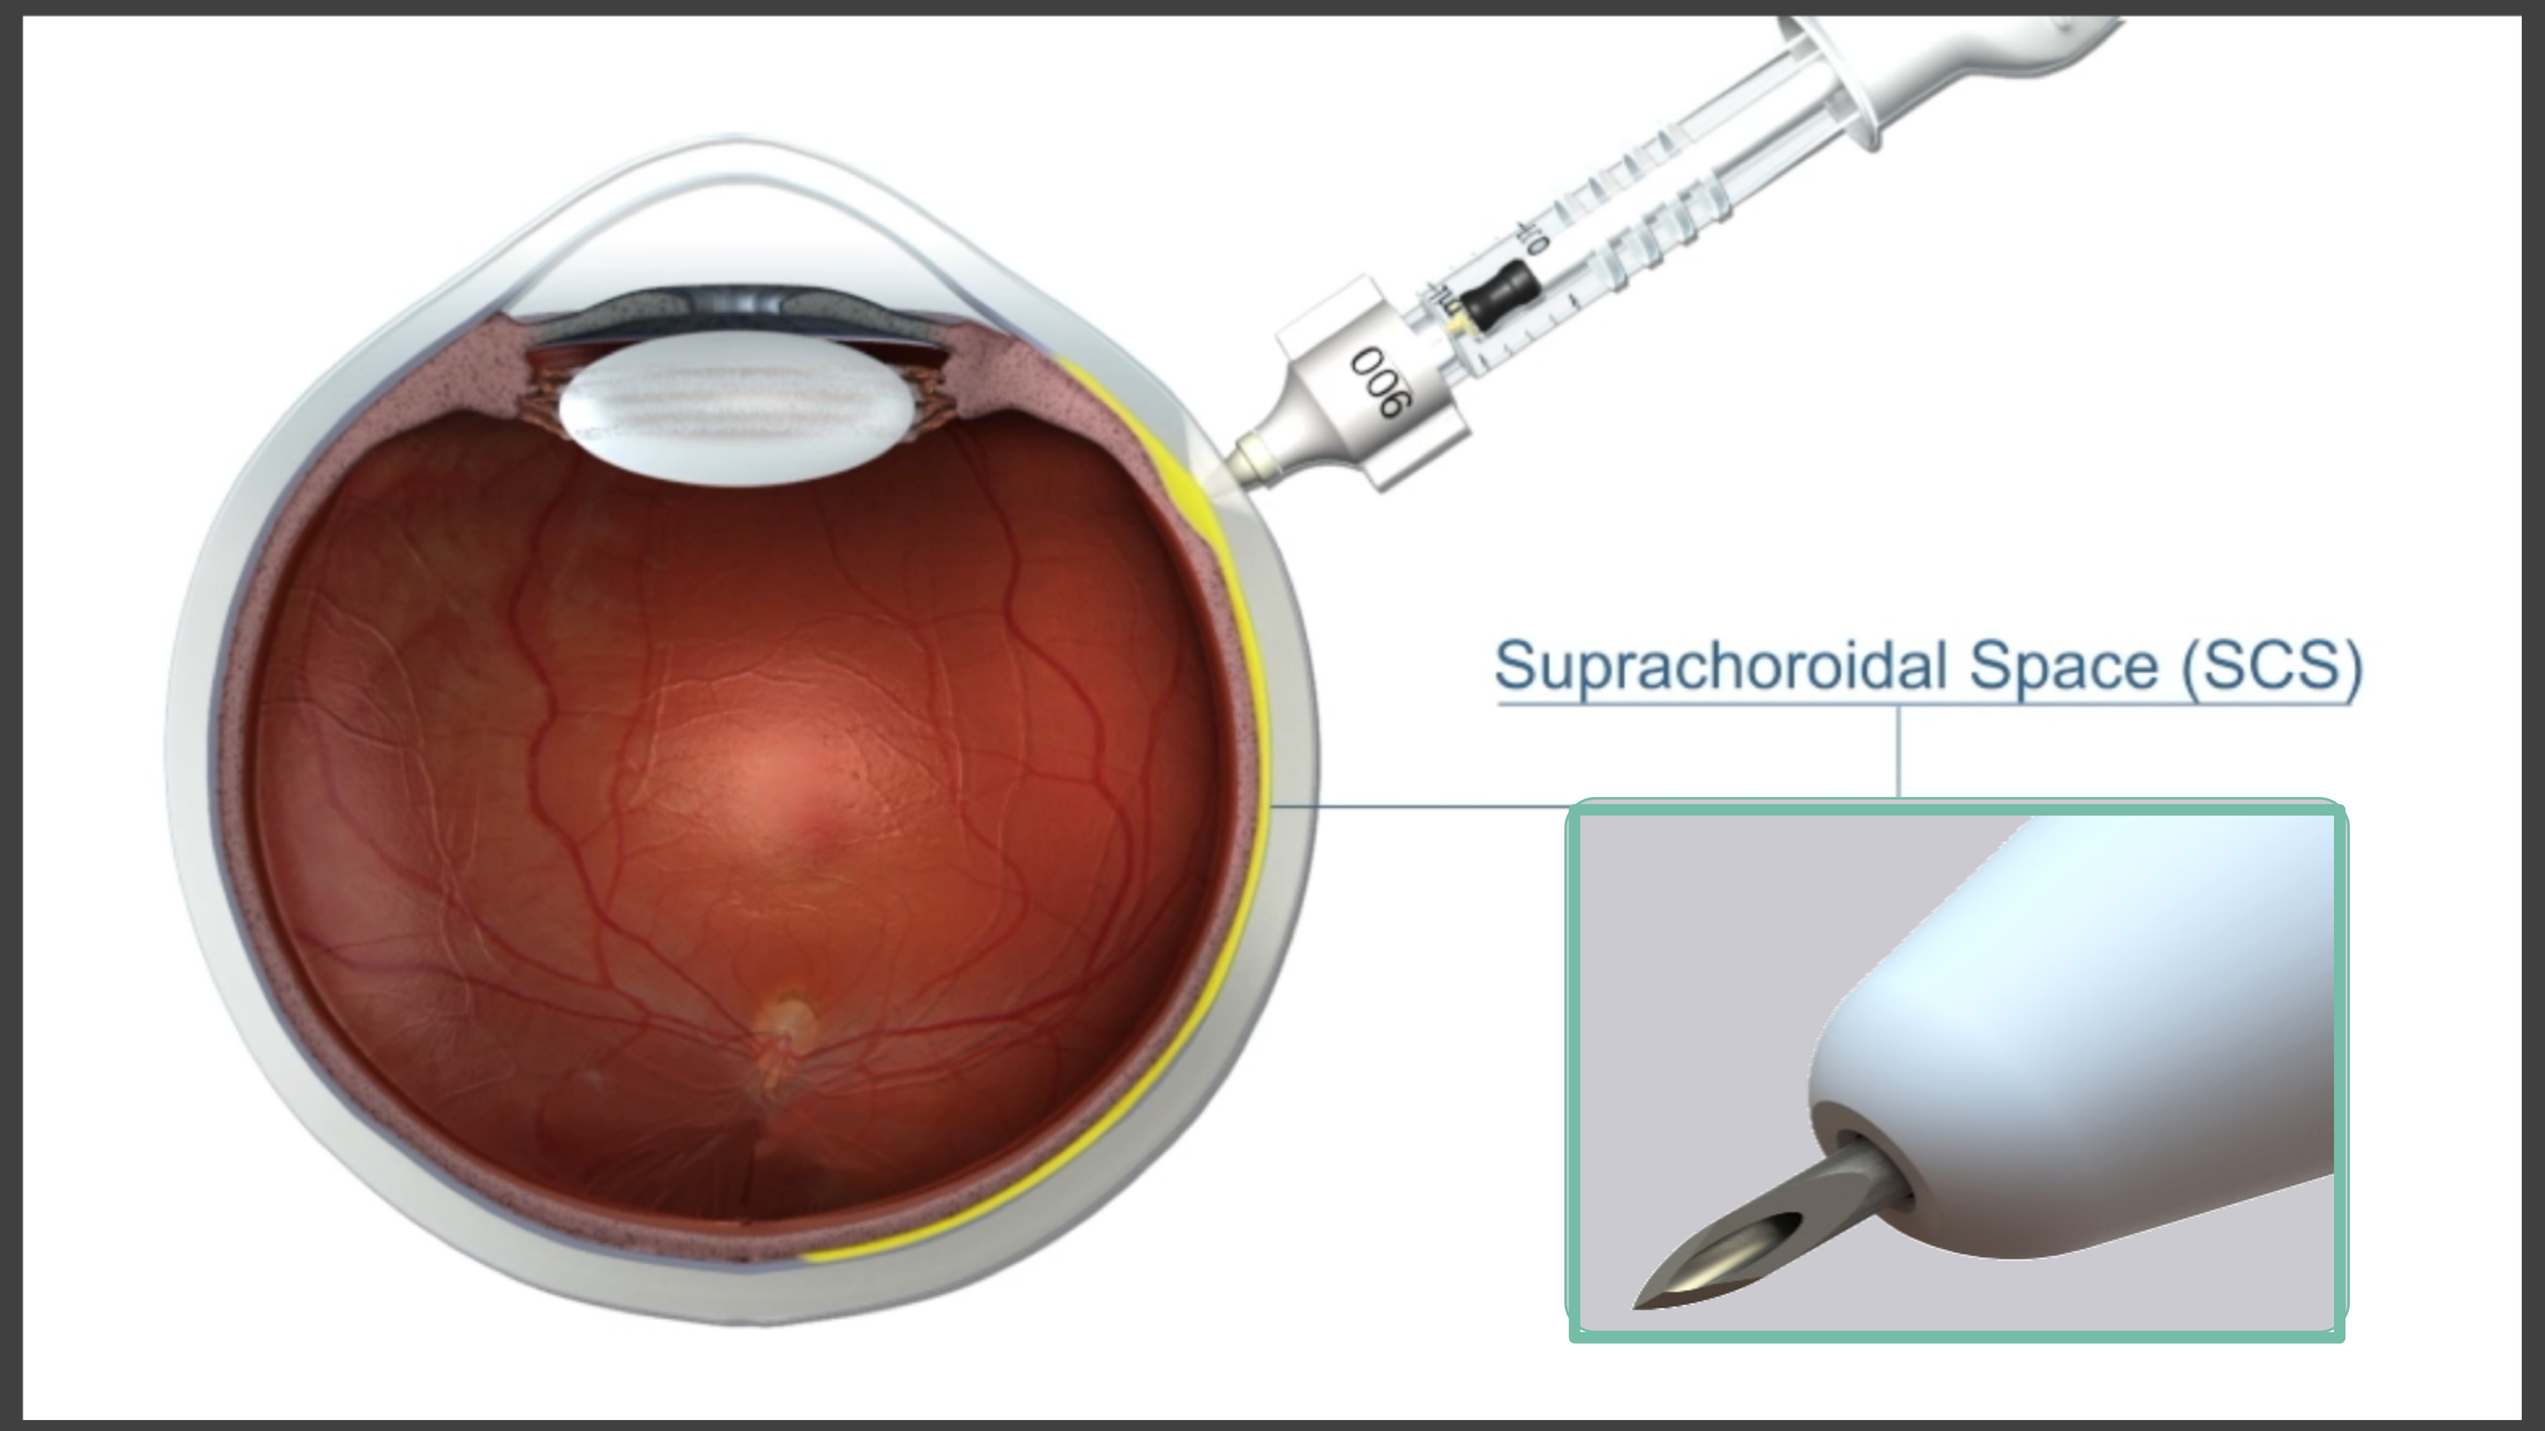
900
Suprachoroidal Space (SCS)

Free length of
900 μm or **1100 μm**

Figure 1: Schematic representation of suprachoroidal CLS-TA delivery

Results

SCI Needle Length Frequency Distributions

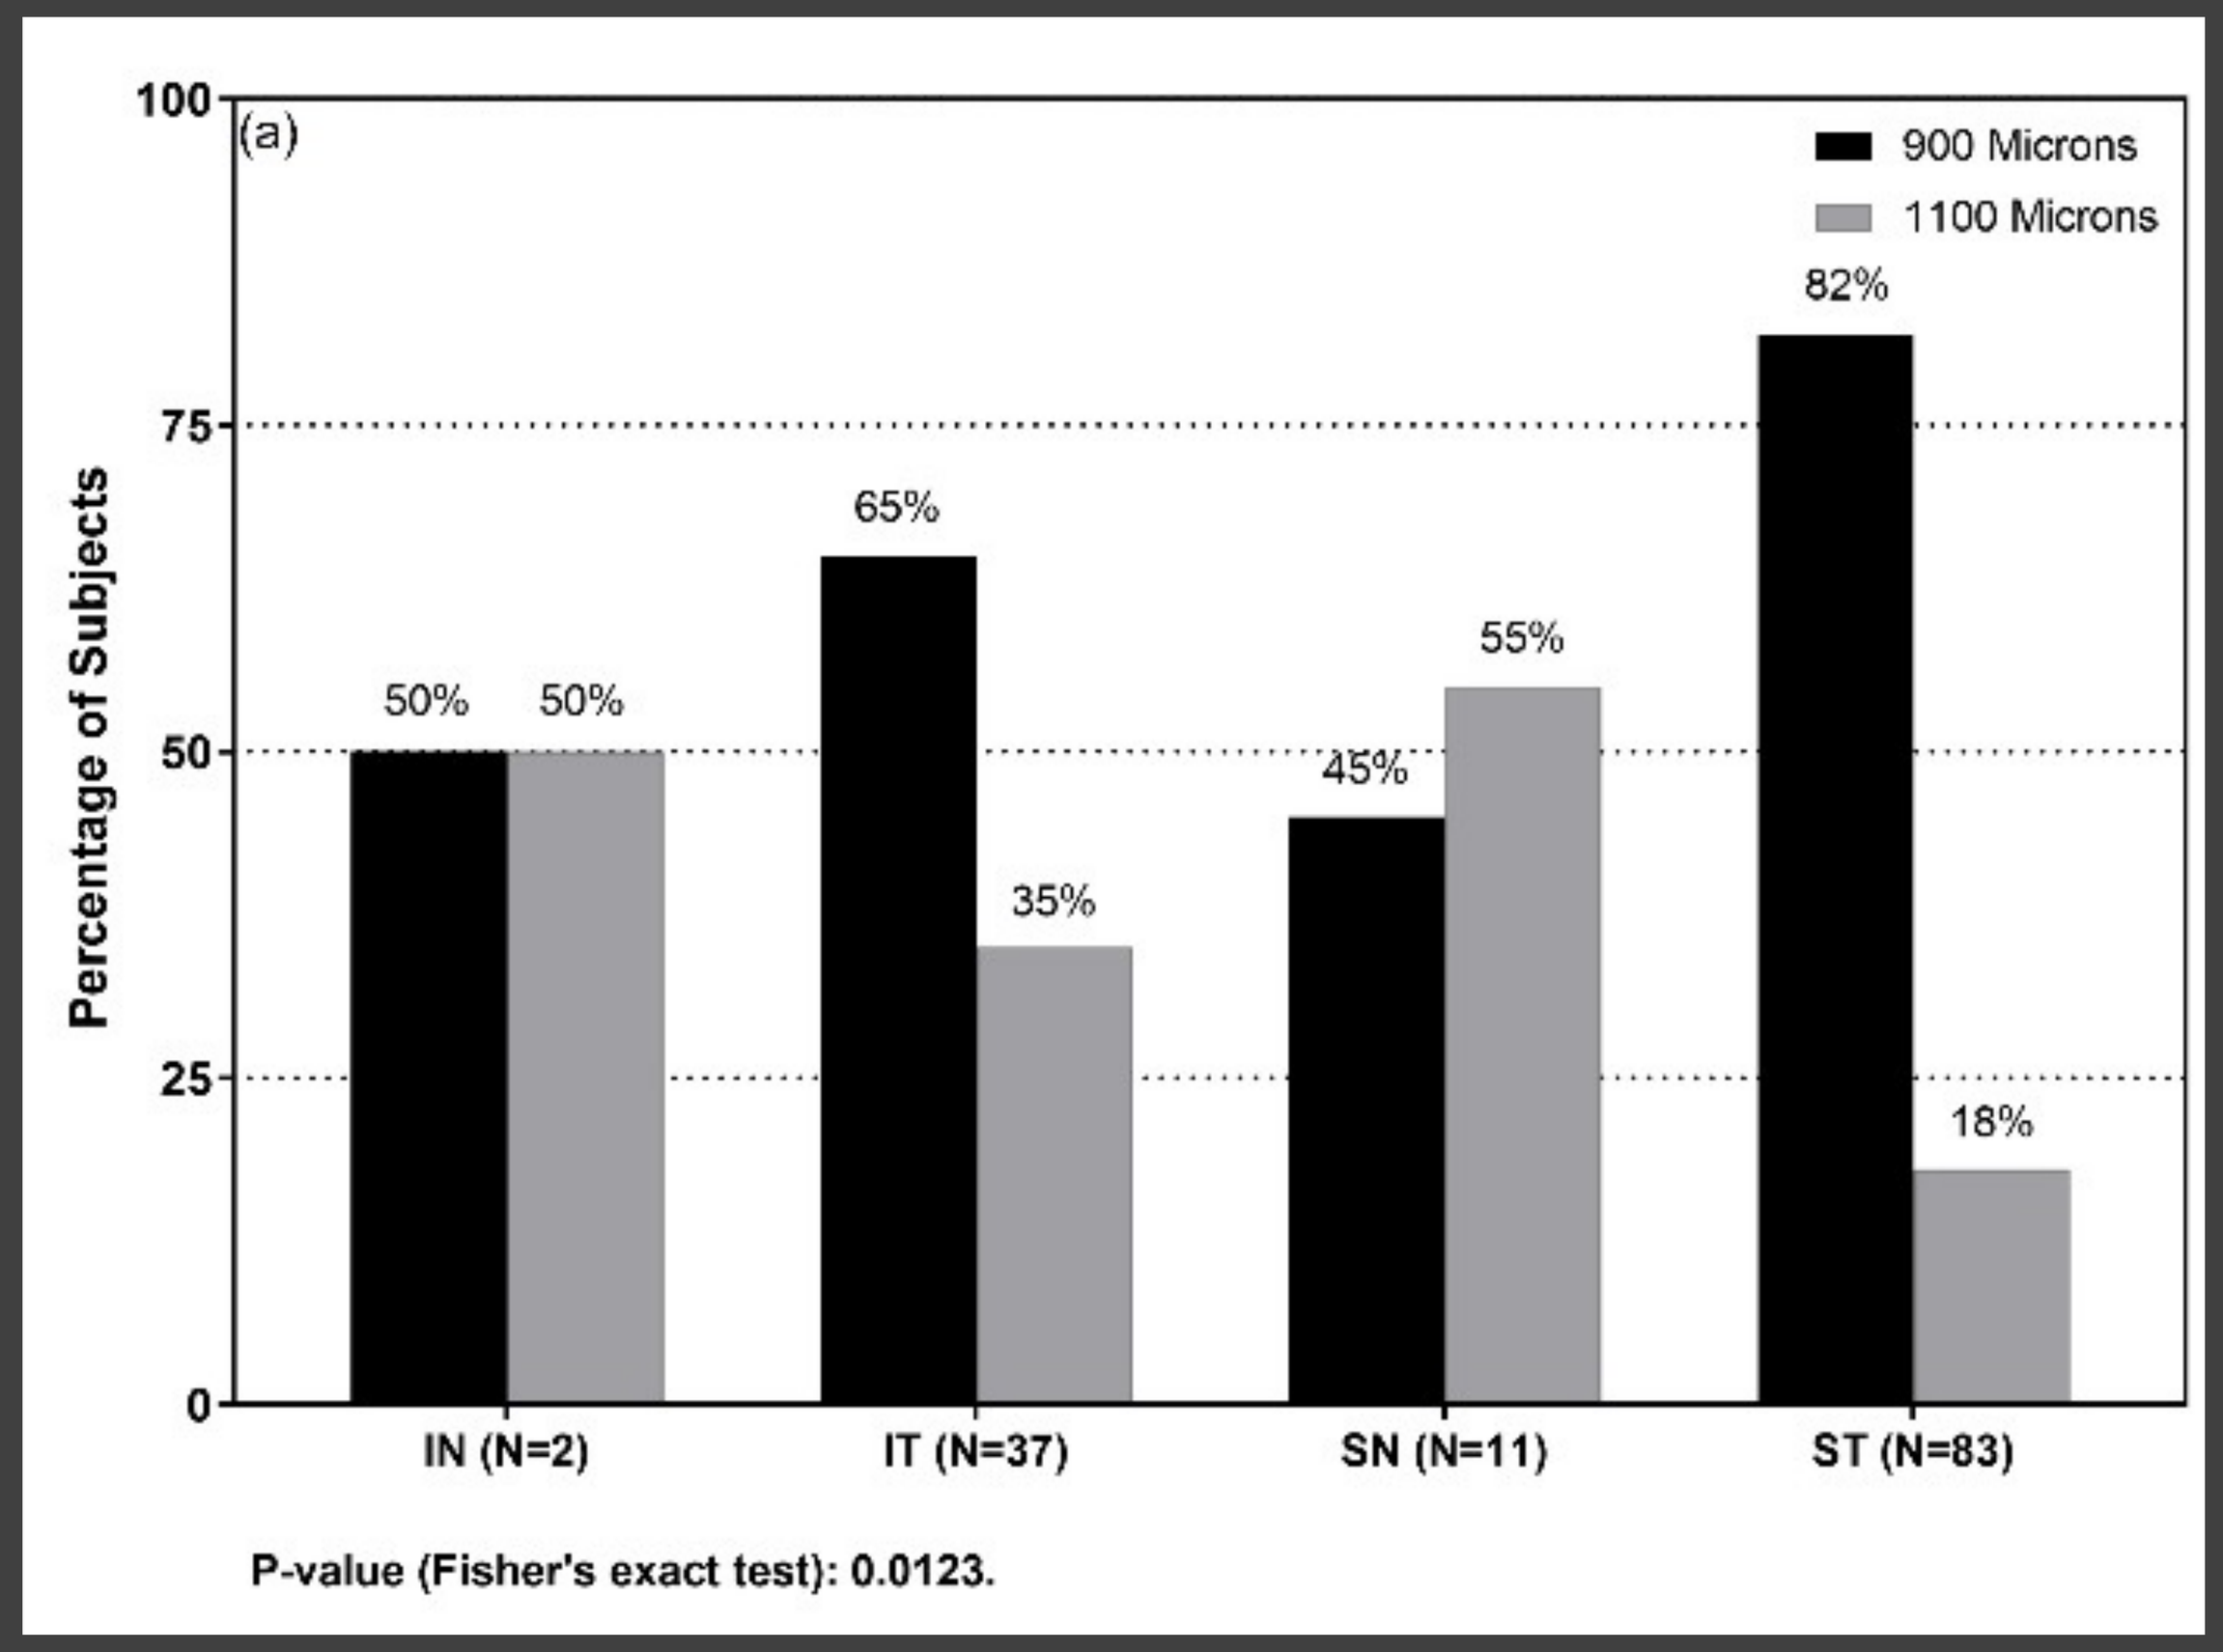
(a)
900 Microns
1100 Microns
Percentage of Subjects
100
75
50
25
0
50% 50%
65%
35%
45%
55%
82%
18%
IN (N=2)
IT (N=37)
SN (N=11)
ST (N=83)
P-value (Fisher's exact test): 0.0123.

Figure 2: Injection Quadrant

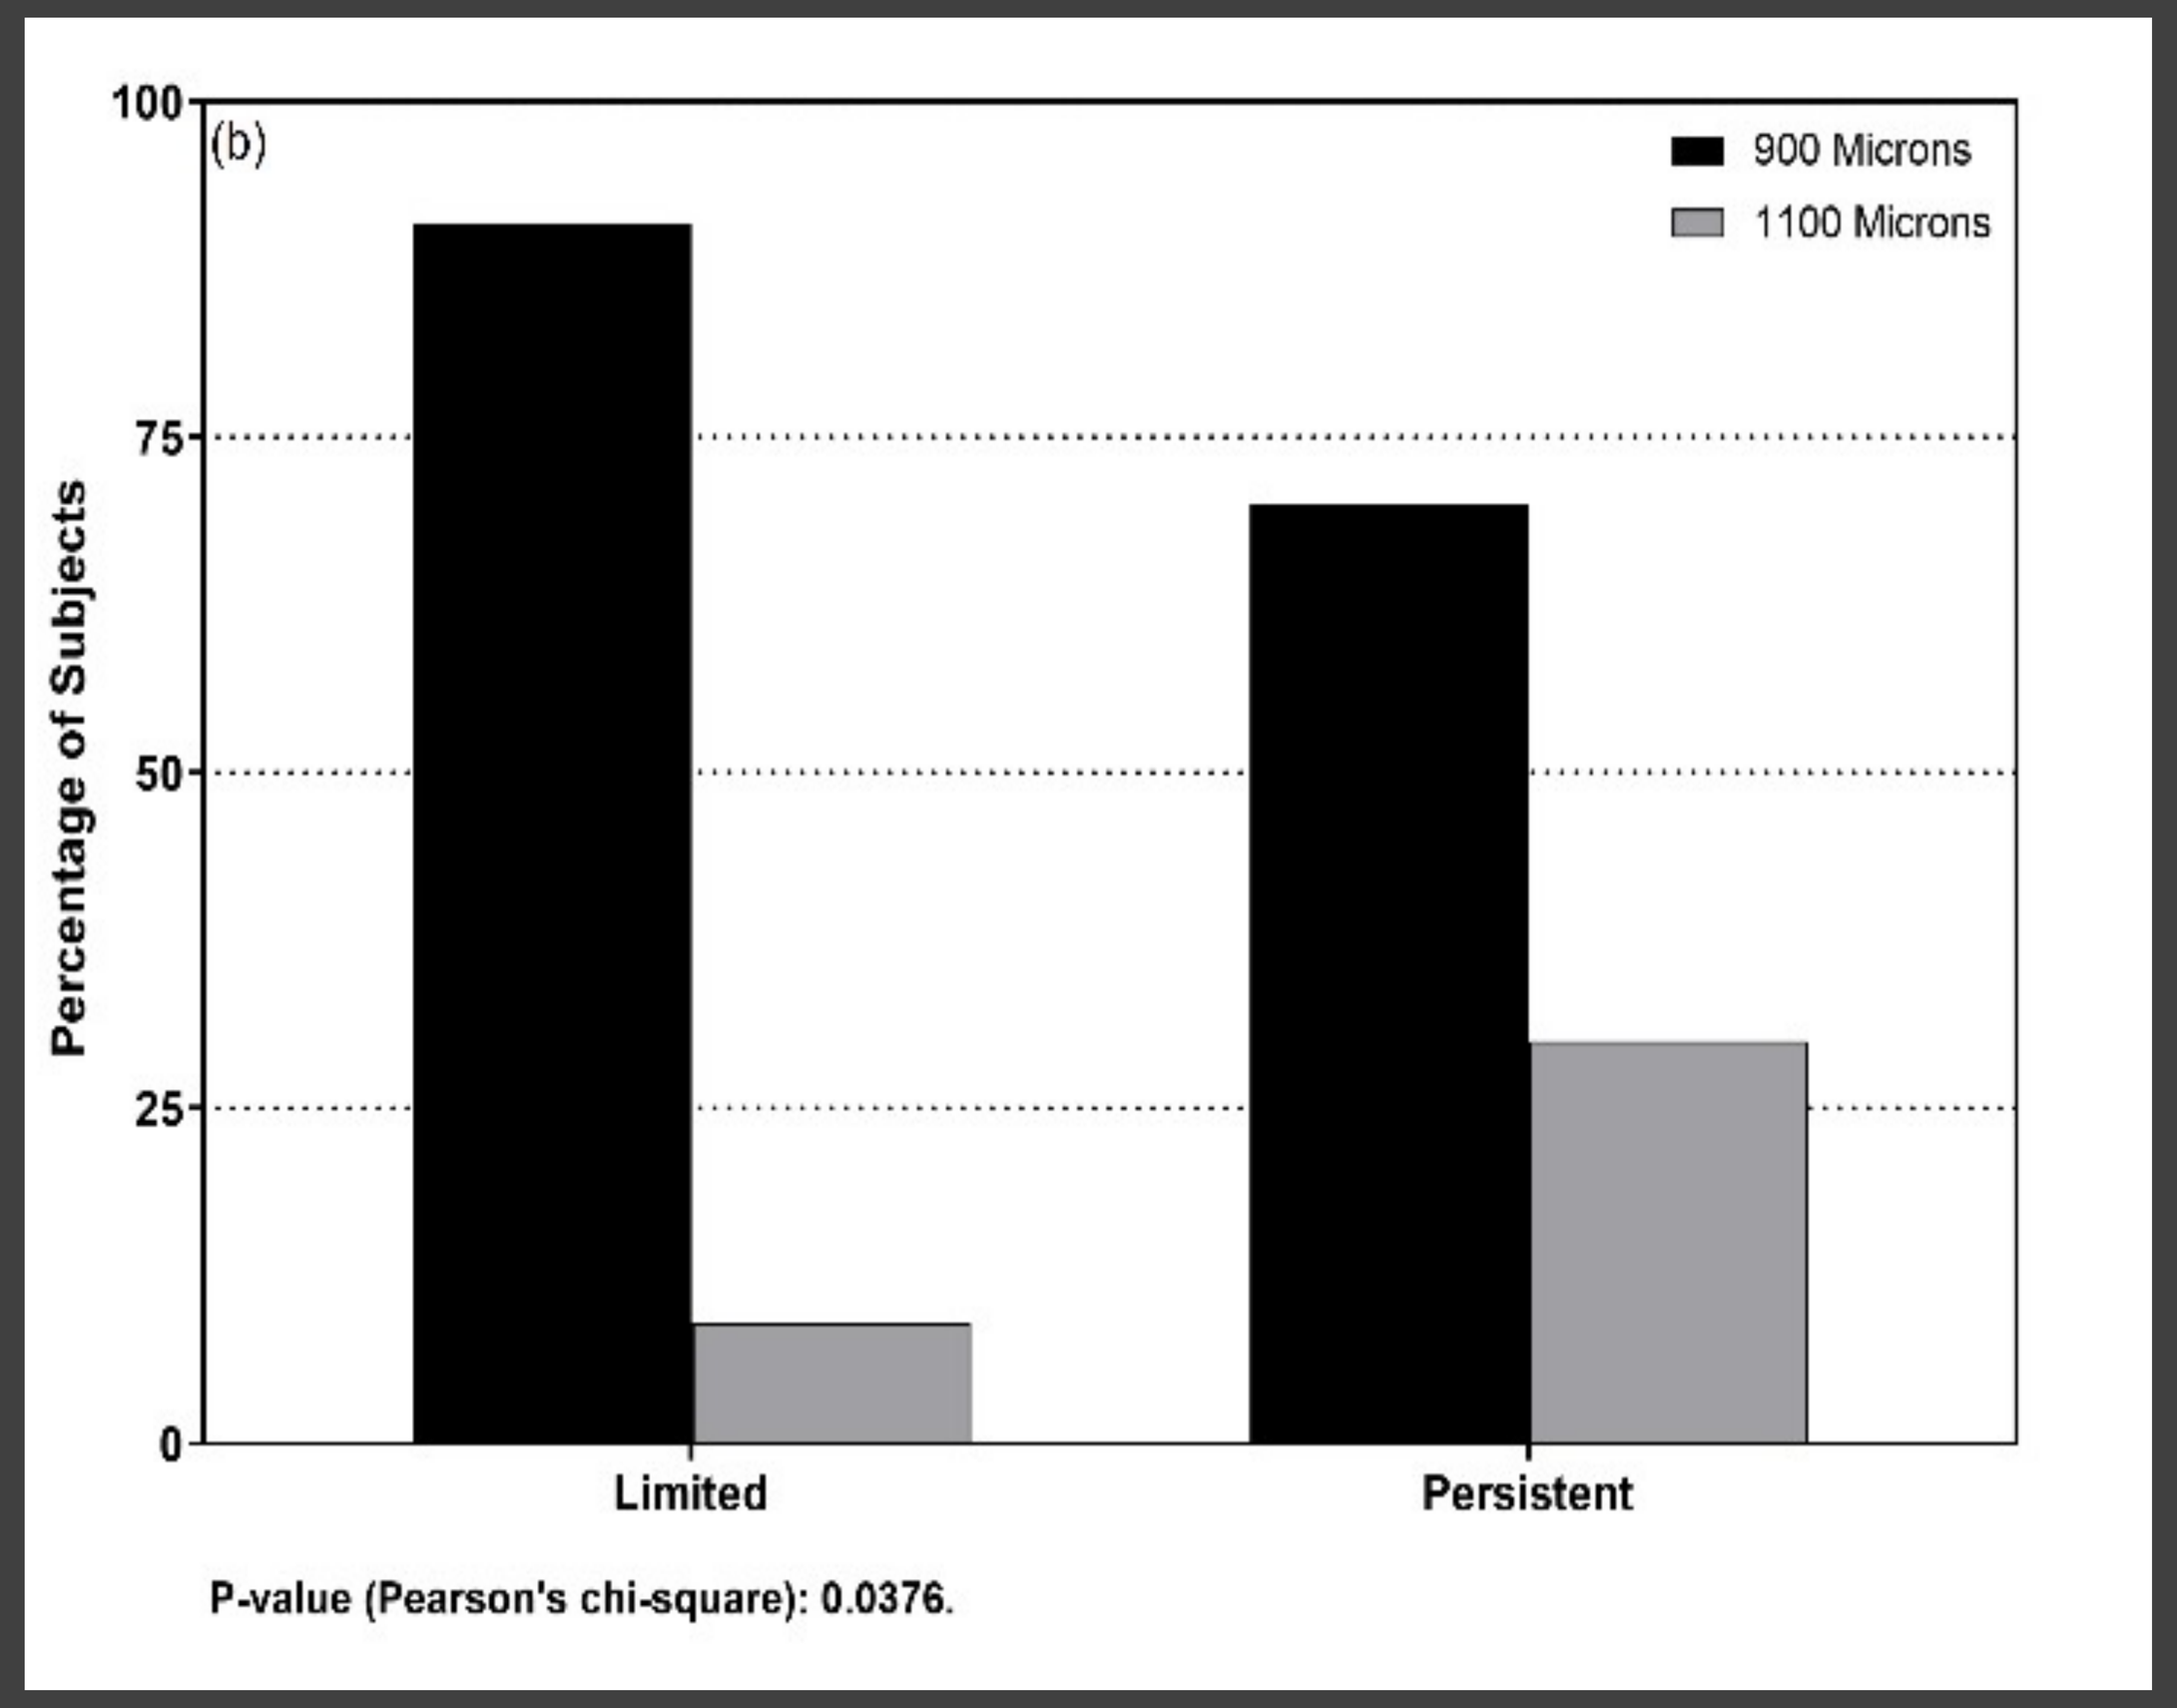
(b)
900 Microns
1100 Microns
Percentage of Subjects
100
75
50
25
0
Limited
Persistent
P-value (Pearson's chi-square): 0.0376.

Figure 3: Uveitis Disease Duration

82% of injections administered superotemporally were completed with the 900 μm needle compared to 45% of injections administered superonasally

91% of injections were completed with the 900 μm needle for Limited (≤ 3 months) and 70% for Persistent (> 3 months)

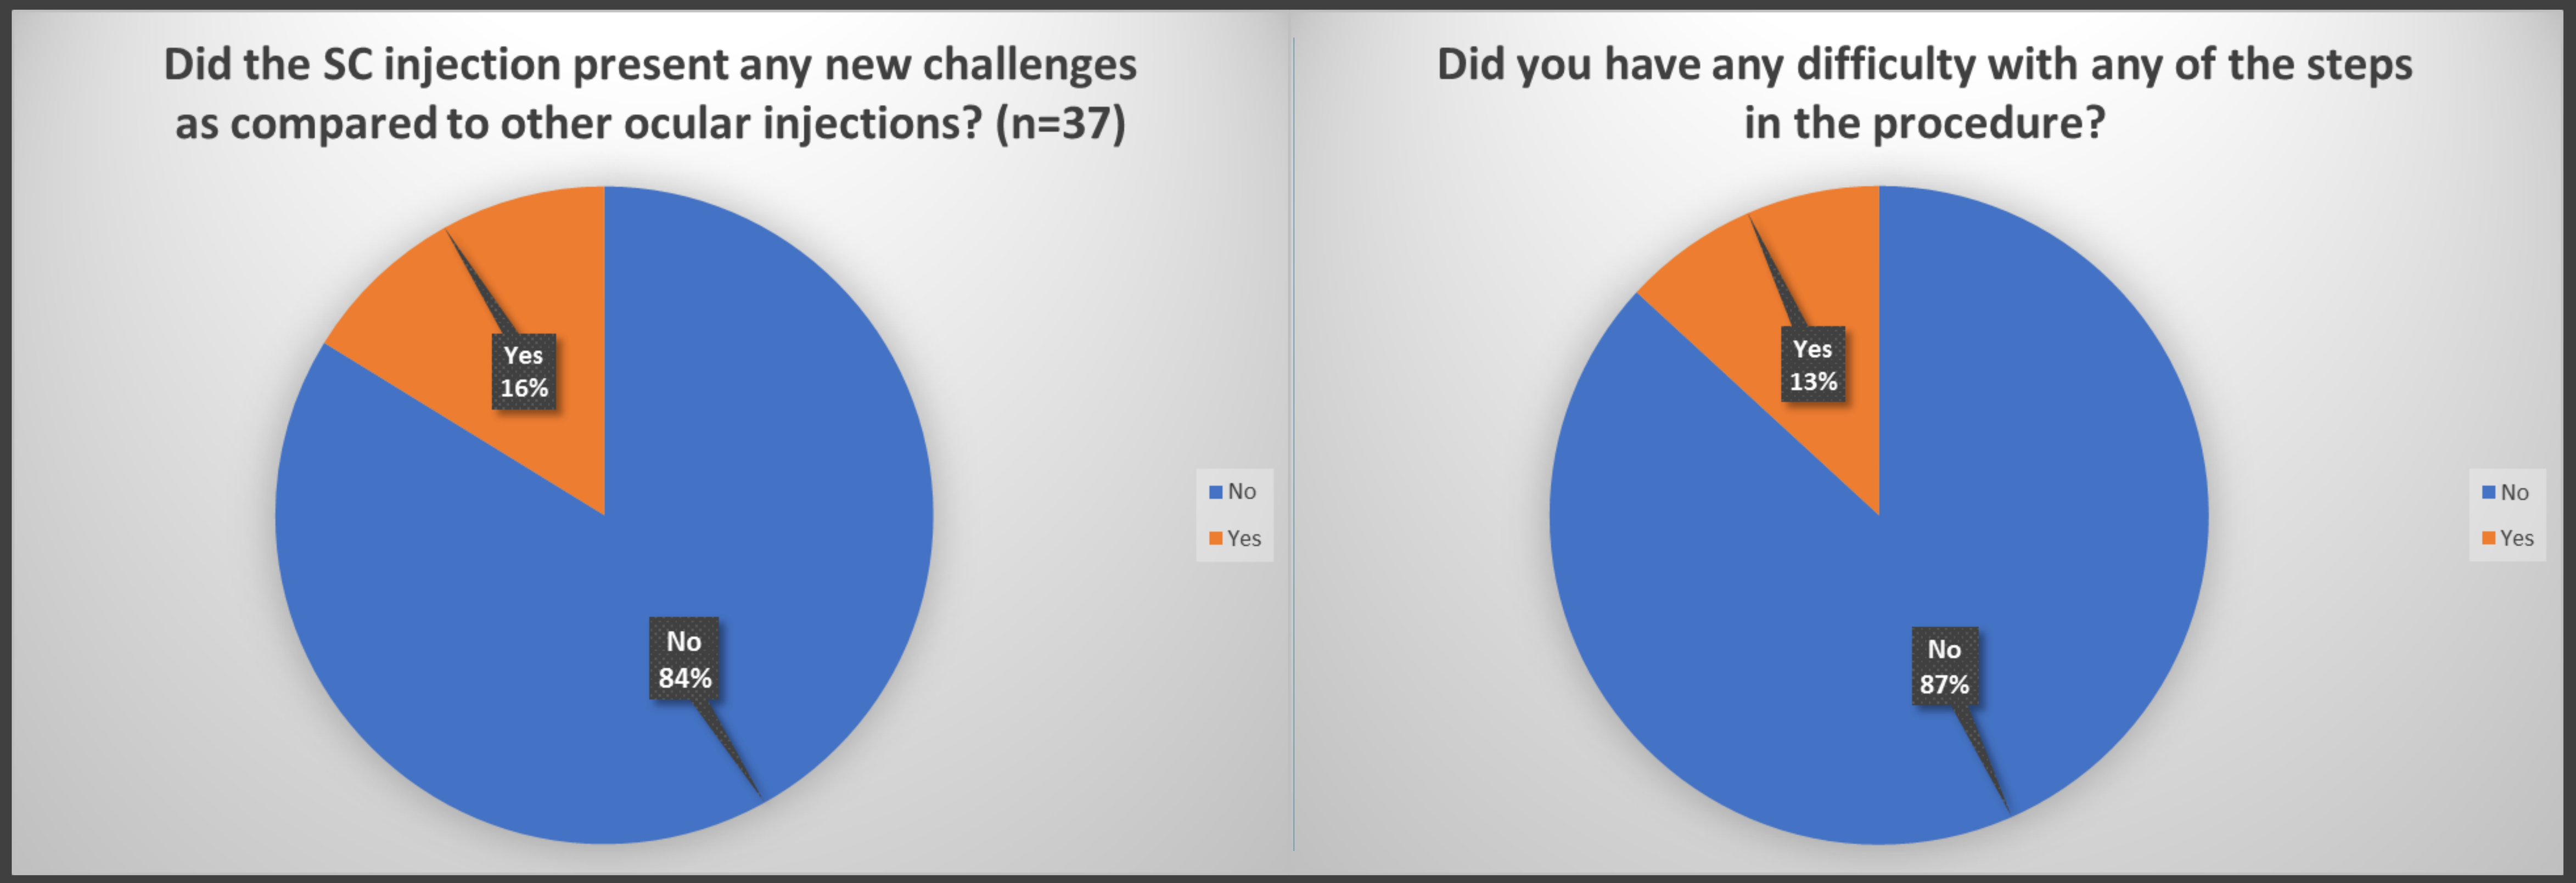
Did the SC injection present any new challenges as compared to other ocular injections? (n=37)
Yes 16%
No 84%
No
Yes
Did you have any difficulty with any of the steps in the procedure?
Yes 13%
No 87%
No
Yes

Figure 3: User Survey Results

In the user experience survey, over 80% of the physicians responded that SCIs presented no new challenges compared to other types of injections.

REFERENCES:

1) Yeh S, Khurana RN, Shah M, et al. Efficacy and safety of suprachoroidal CLS-TA for macular edema secondary to noninfectious uveitis: phase 3, randomized trial. Ophthalmology. 2020;127(7):948–955.

2) Lampen SIR, Khurana RN, Noronha G, Brown DM, Wykoff CC. Suprachoroidal space alterations following delivery of triamcinolone acetonide: Post-hoc analysis of the phase 1/2 HULK study of patients with diabetic macular edema. Ophthalmic Surg Lasers Imaging Retina. 2018;49(9):692– 697.

3) Henry CR, Shah M, Barakat MR, Dayani P, Wang RC, Khurana RN, Rifkin L, Yeh S, Hall C, Ciulla T. Suprachoroidal CLS-TA for non-infectious uveitis: an open-label, safety trial (AZALEA). Br J Ophthalmol. 2021 Feb 5:

- Of the133 total baseline SCIs,
 - 74% were successfully completed with the 900μm needle.
 - remaining 26% with the 1100μm needle.
- Univariate analysis revealed no relationship between needle length utilized, gender, lens status, uveitis sub-type, disease course or onset.
- Multivariate logistical regression verified univariate analysis demonstrating the potential impact of age, disease duration and SCI quadrant on needle length.
- <u>**Correlations**</u>
 - Disease duration was statistically correlated with needle length:
 - 91% of injections were completed with the 900μm needle for Limited (≤ 3 months) and 70% for Persistent (> 3 months).
 - Age was moderately inversely correlated with needle length
 - Injection quadrant was statistically related:
 - 82% of injections administered superotemporally were completed with the 900μm needle compared to 45% of injections administered superonasally.

Conclusions

- Study analyses are limited by retrospective nature and relatively small sample size
- However, few patient characteristics were found to correlate with needle length, indicating the procedure can be completed for the majority of patients with the 900μm needle
- SCIs with the SCS Microinjector have potential to reliably and repeatably deliver drugs for chorioretinal diseases in an in-office setting.
- Survey results suggest that most physicians doing intravitreal treatments will be able to transition to SCIs relatively easily