C0234

A Novel Technique to Characterize Key Fluid Mechanic Properties of the Suprachoroidal Injection Procedure in an *In Vivo* Model

Nathan Fisher, Jesse Yoo, Shelley Hancock, Rafael V. Andino — Clearside Biomedical, Inc., Atlanta, GA

Purpose

- This study was conducted to characterize and evaluate the fluid mechanics resulting from suprachoroidal injections in an *in vivo* pig model.

Methods

- CLS-TA (triamcinolone acetonide injectable suspension), 40 mg/mL, was administered suprachoroidally to pig eyes at a volume of 100 µL per injection for the evaluation of real-time force and in-line pressure during the procedure. Both eyes on 2 separate specimens were each injected twice for a total of 8 suprachoroidal injections. Subsequent injections in the same eye were performed in opposing hemispheres, and a recovery period was allowed for the bolus of injected drug to disperse throughout the suprachoroidal space and for the intraocular pressure to equilibrate. The proprietary device for the suprachoroidal injection—an SCS Microinjector—is a manual piston syringe with a plunger that is encased in a handle, a unique needle hub design, and a needle that is ~1000 µm in length. The instrumentation for the evaluations described included a force transducer located within the handle (plunger) of the syringe and an in-line pressure sensor integrated into the fluid path between the needle and the syringe (Figure 1). Additionally, a Computational Fluid Dynamics (CFD) model was developed to characterize the pressure gradients within the microinjector and suprachoroidal space of the eye at the time of injection.

Figure 1. SCS Microinjector With Instrumentation

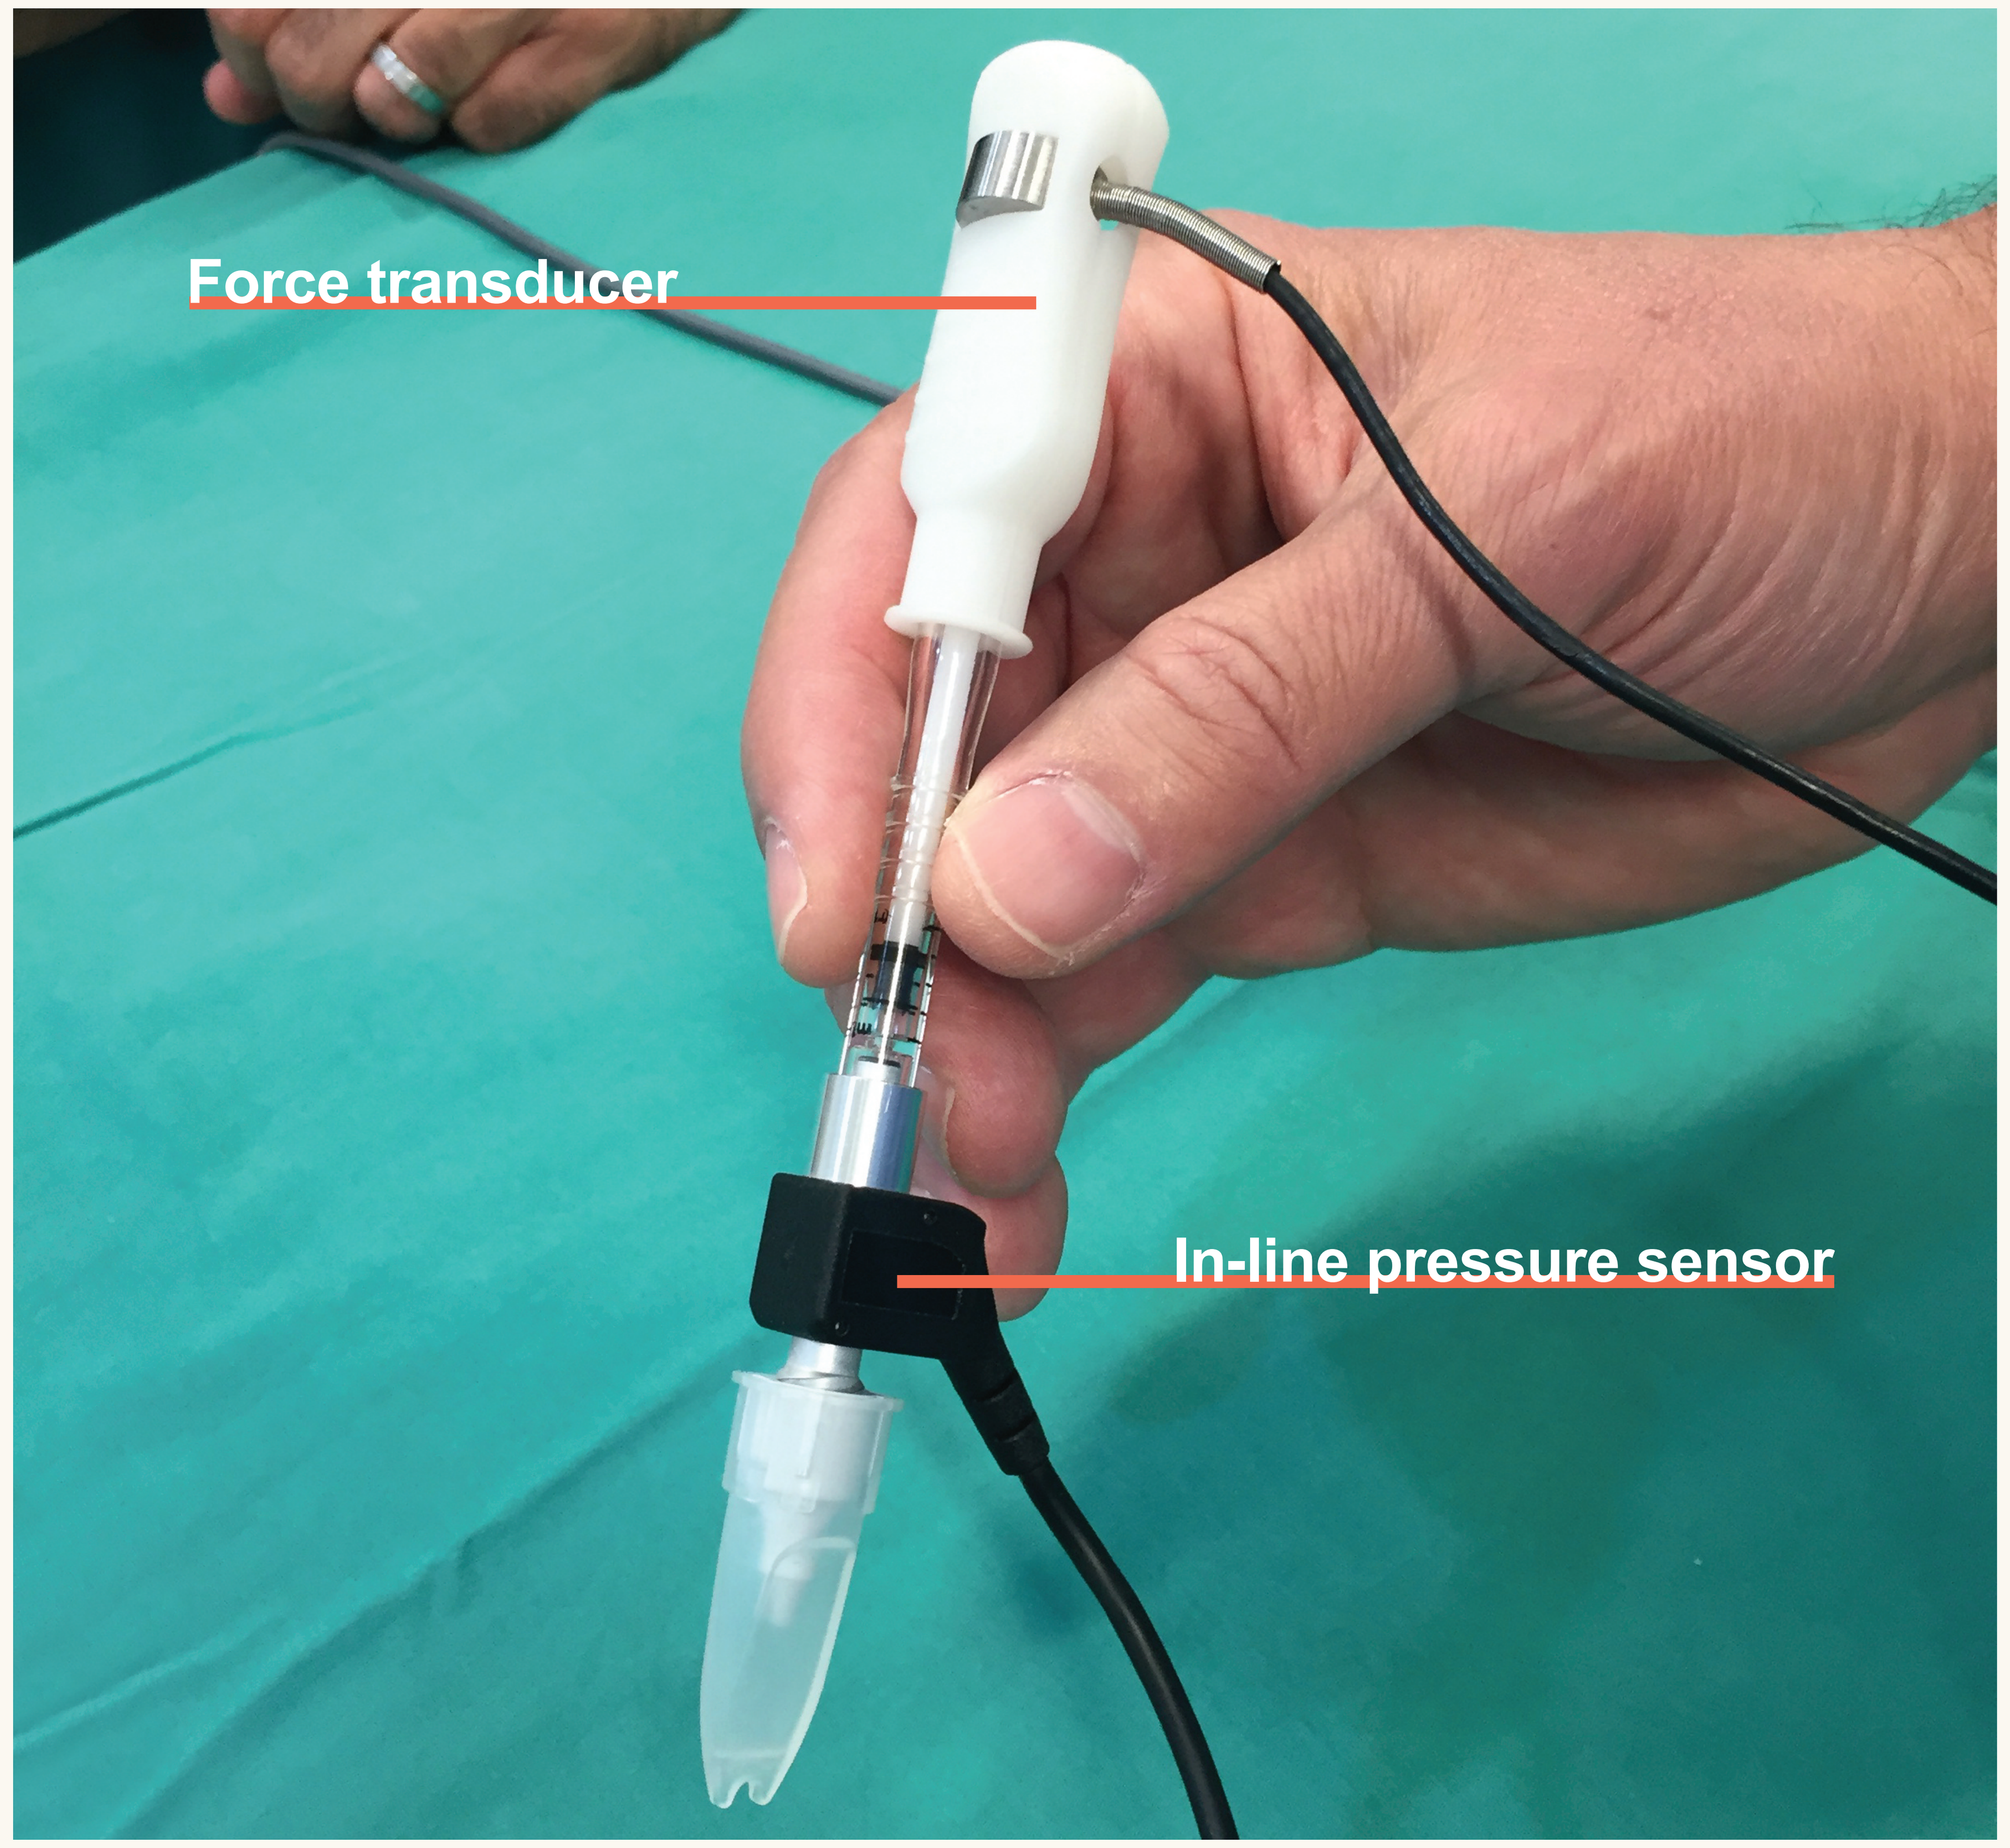
Force transducer
In-line pressure sensor

Results

- The results from the suprachoroidal injections into the pig eyes yielded a range of in-line pressures between 8.85 and 18.41 psi (61.02 and 126.93 kPa) when injection forces of 0.33–0.64 lbf (1.47–2.85 N) were applied to the handle of the microinjector syringe to initiate the suprachoroidal injections (Figure 2). The results from the CFD model displayed a substantial difference between in-line pressure of the syringe and pressure at the point of injection, the needle orifice.

Figure 2. Force and Pressure During Injection

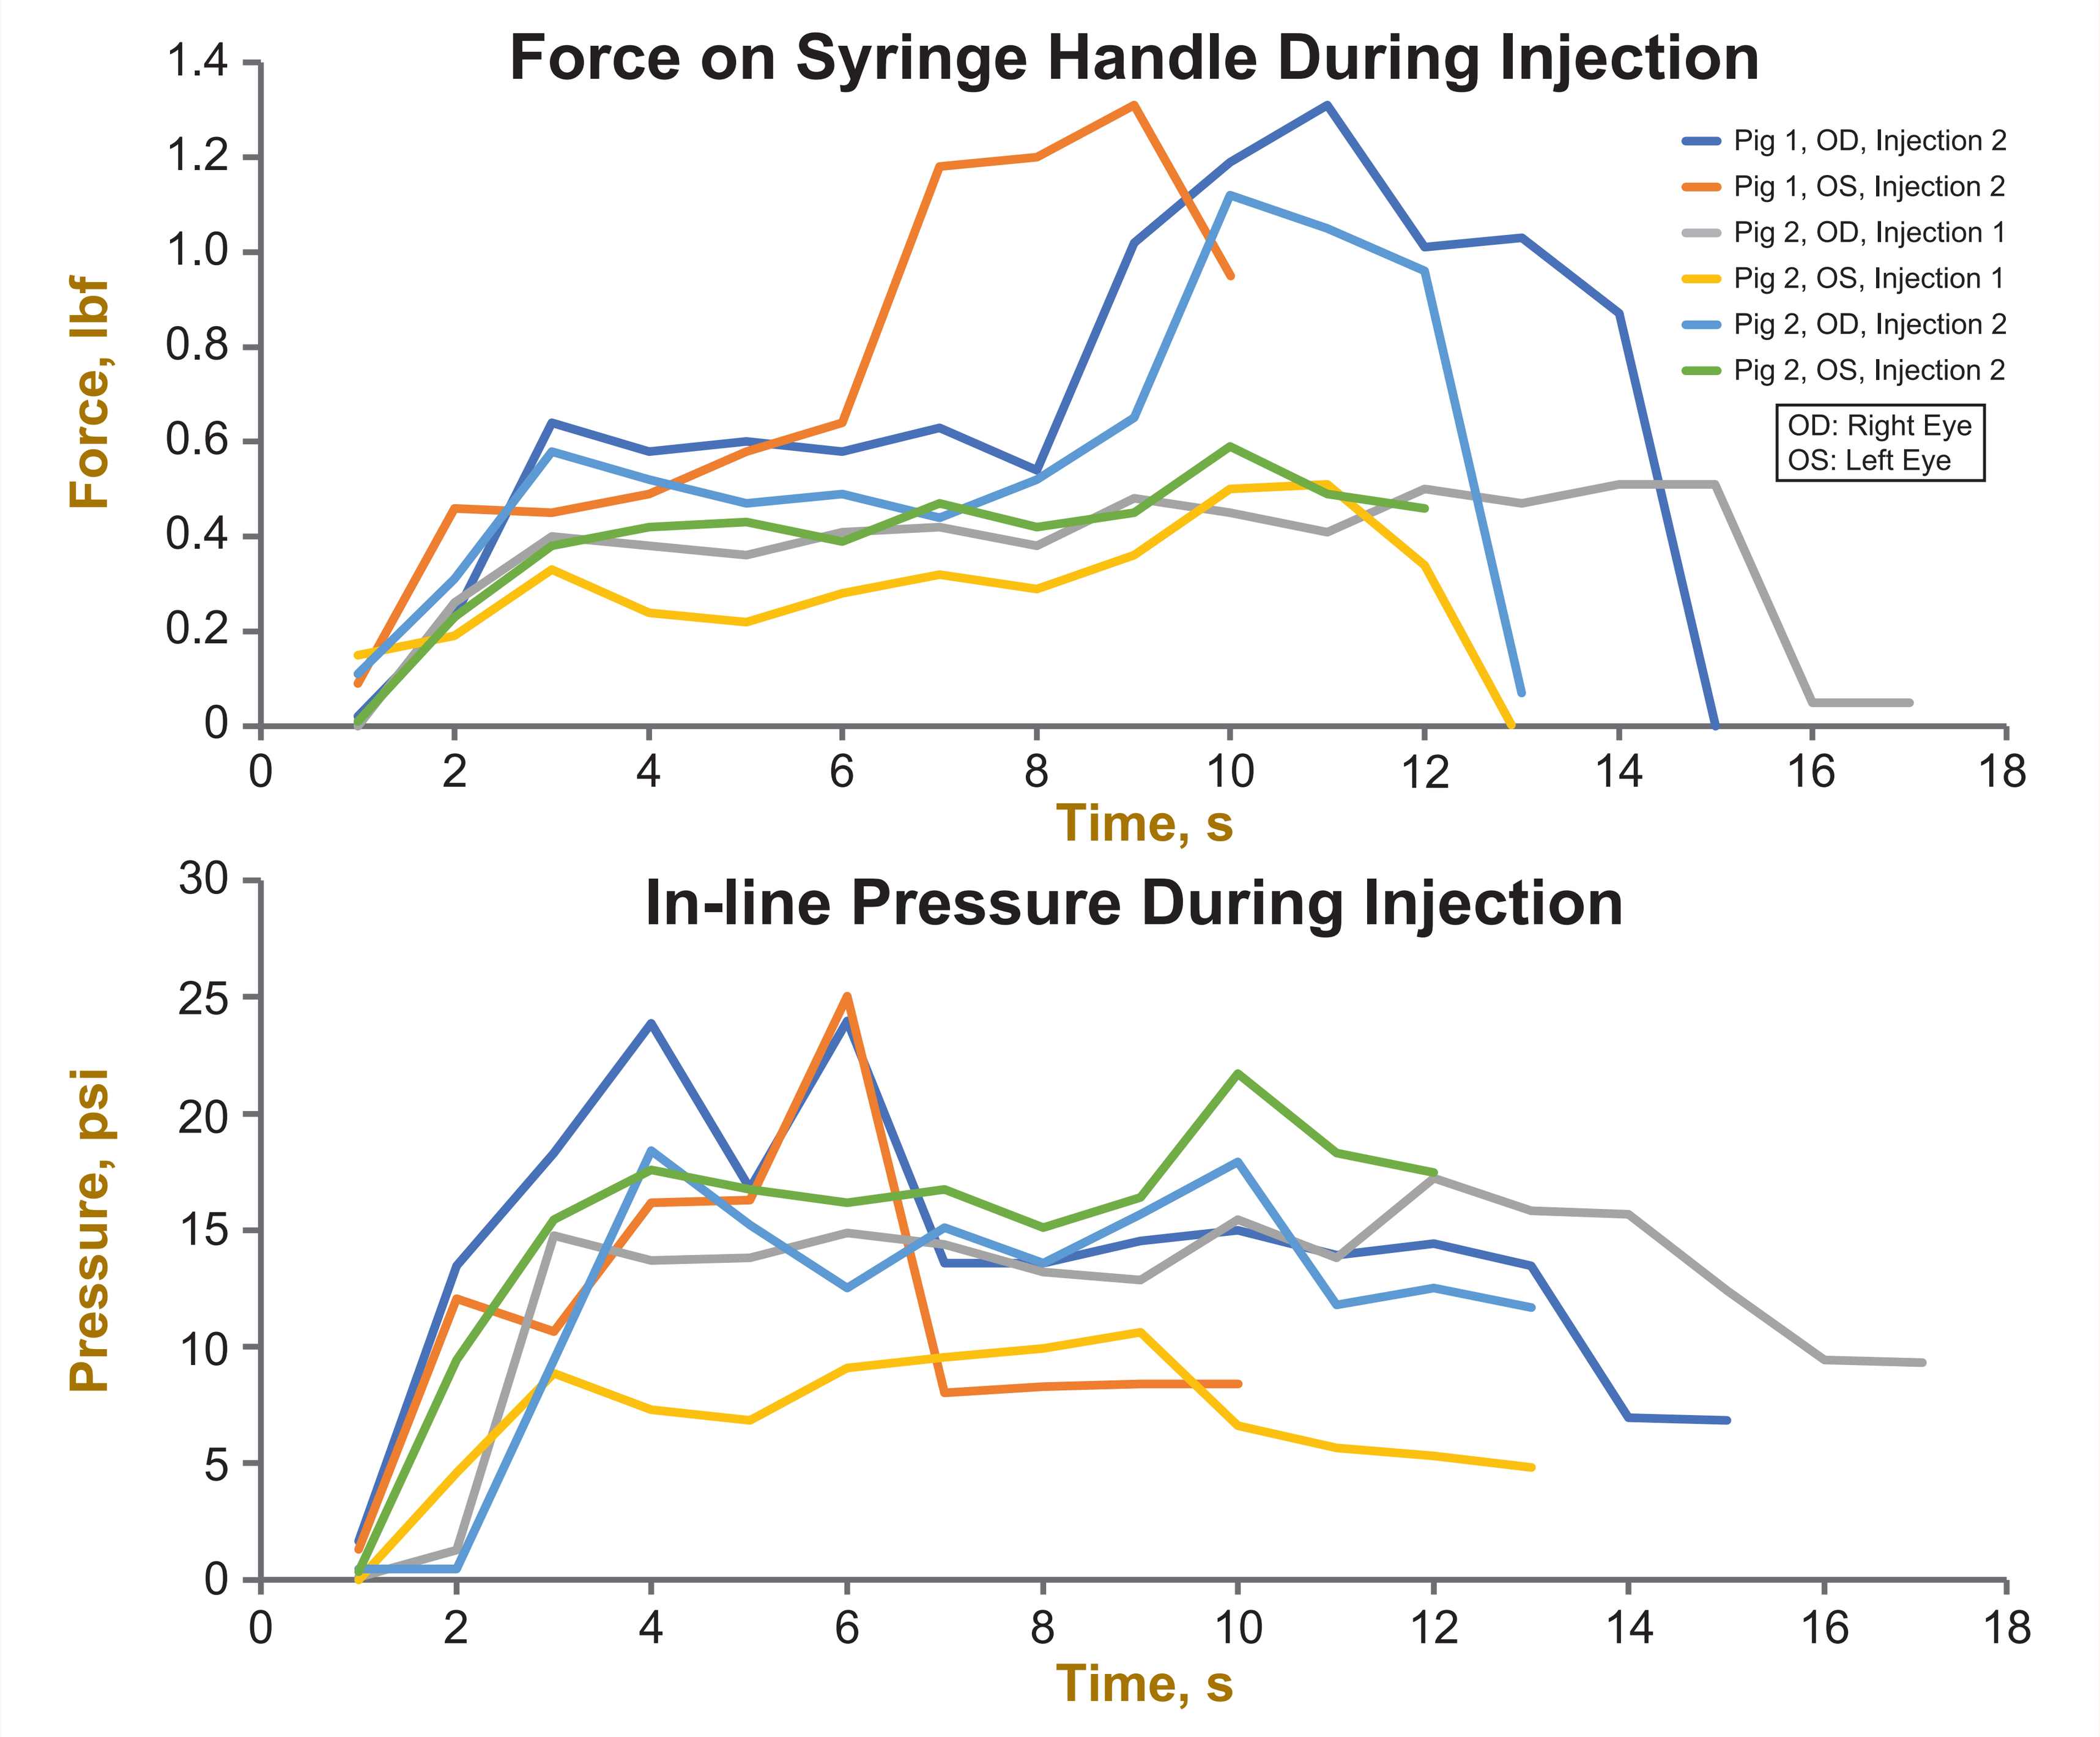
Force on Syringe Handle During Injection
Force, lbf
Time, s
Pig 1, OD, Injection 2
Pig 1, OS, Injection 2
Pig 2, OD, Injection 1
Pig 2, OS, Injection 1
Pig 2, OD, Injection 2
Pig 2, OS, Injection 2
OD: Right Eye
OS: Left Eye
In-line Pressure During Injection
Pressure, psi
Time, s

Figure 3. Force and Pressure to Initiate Injection

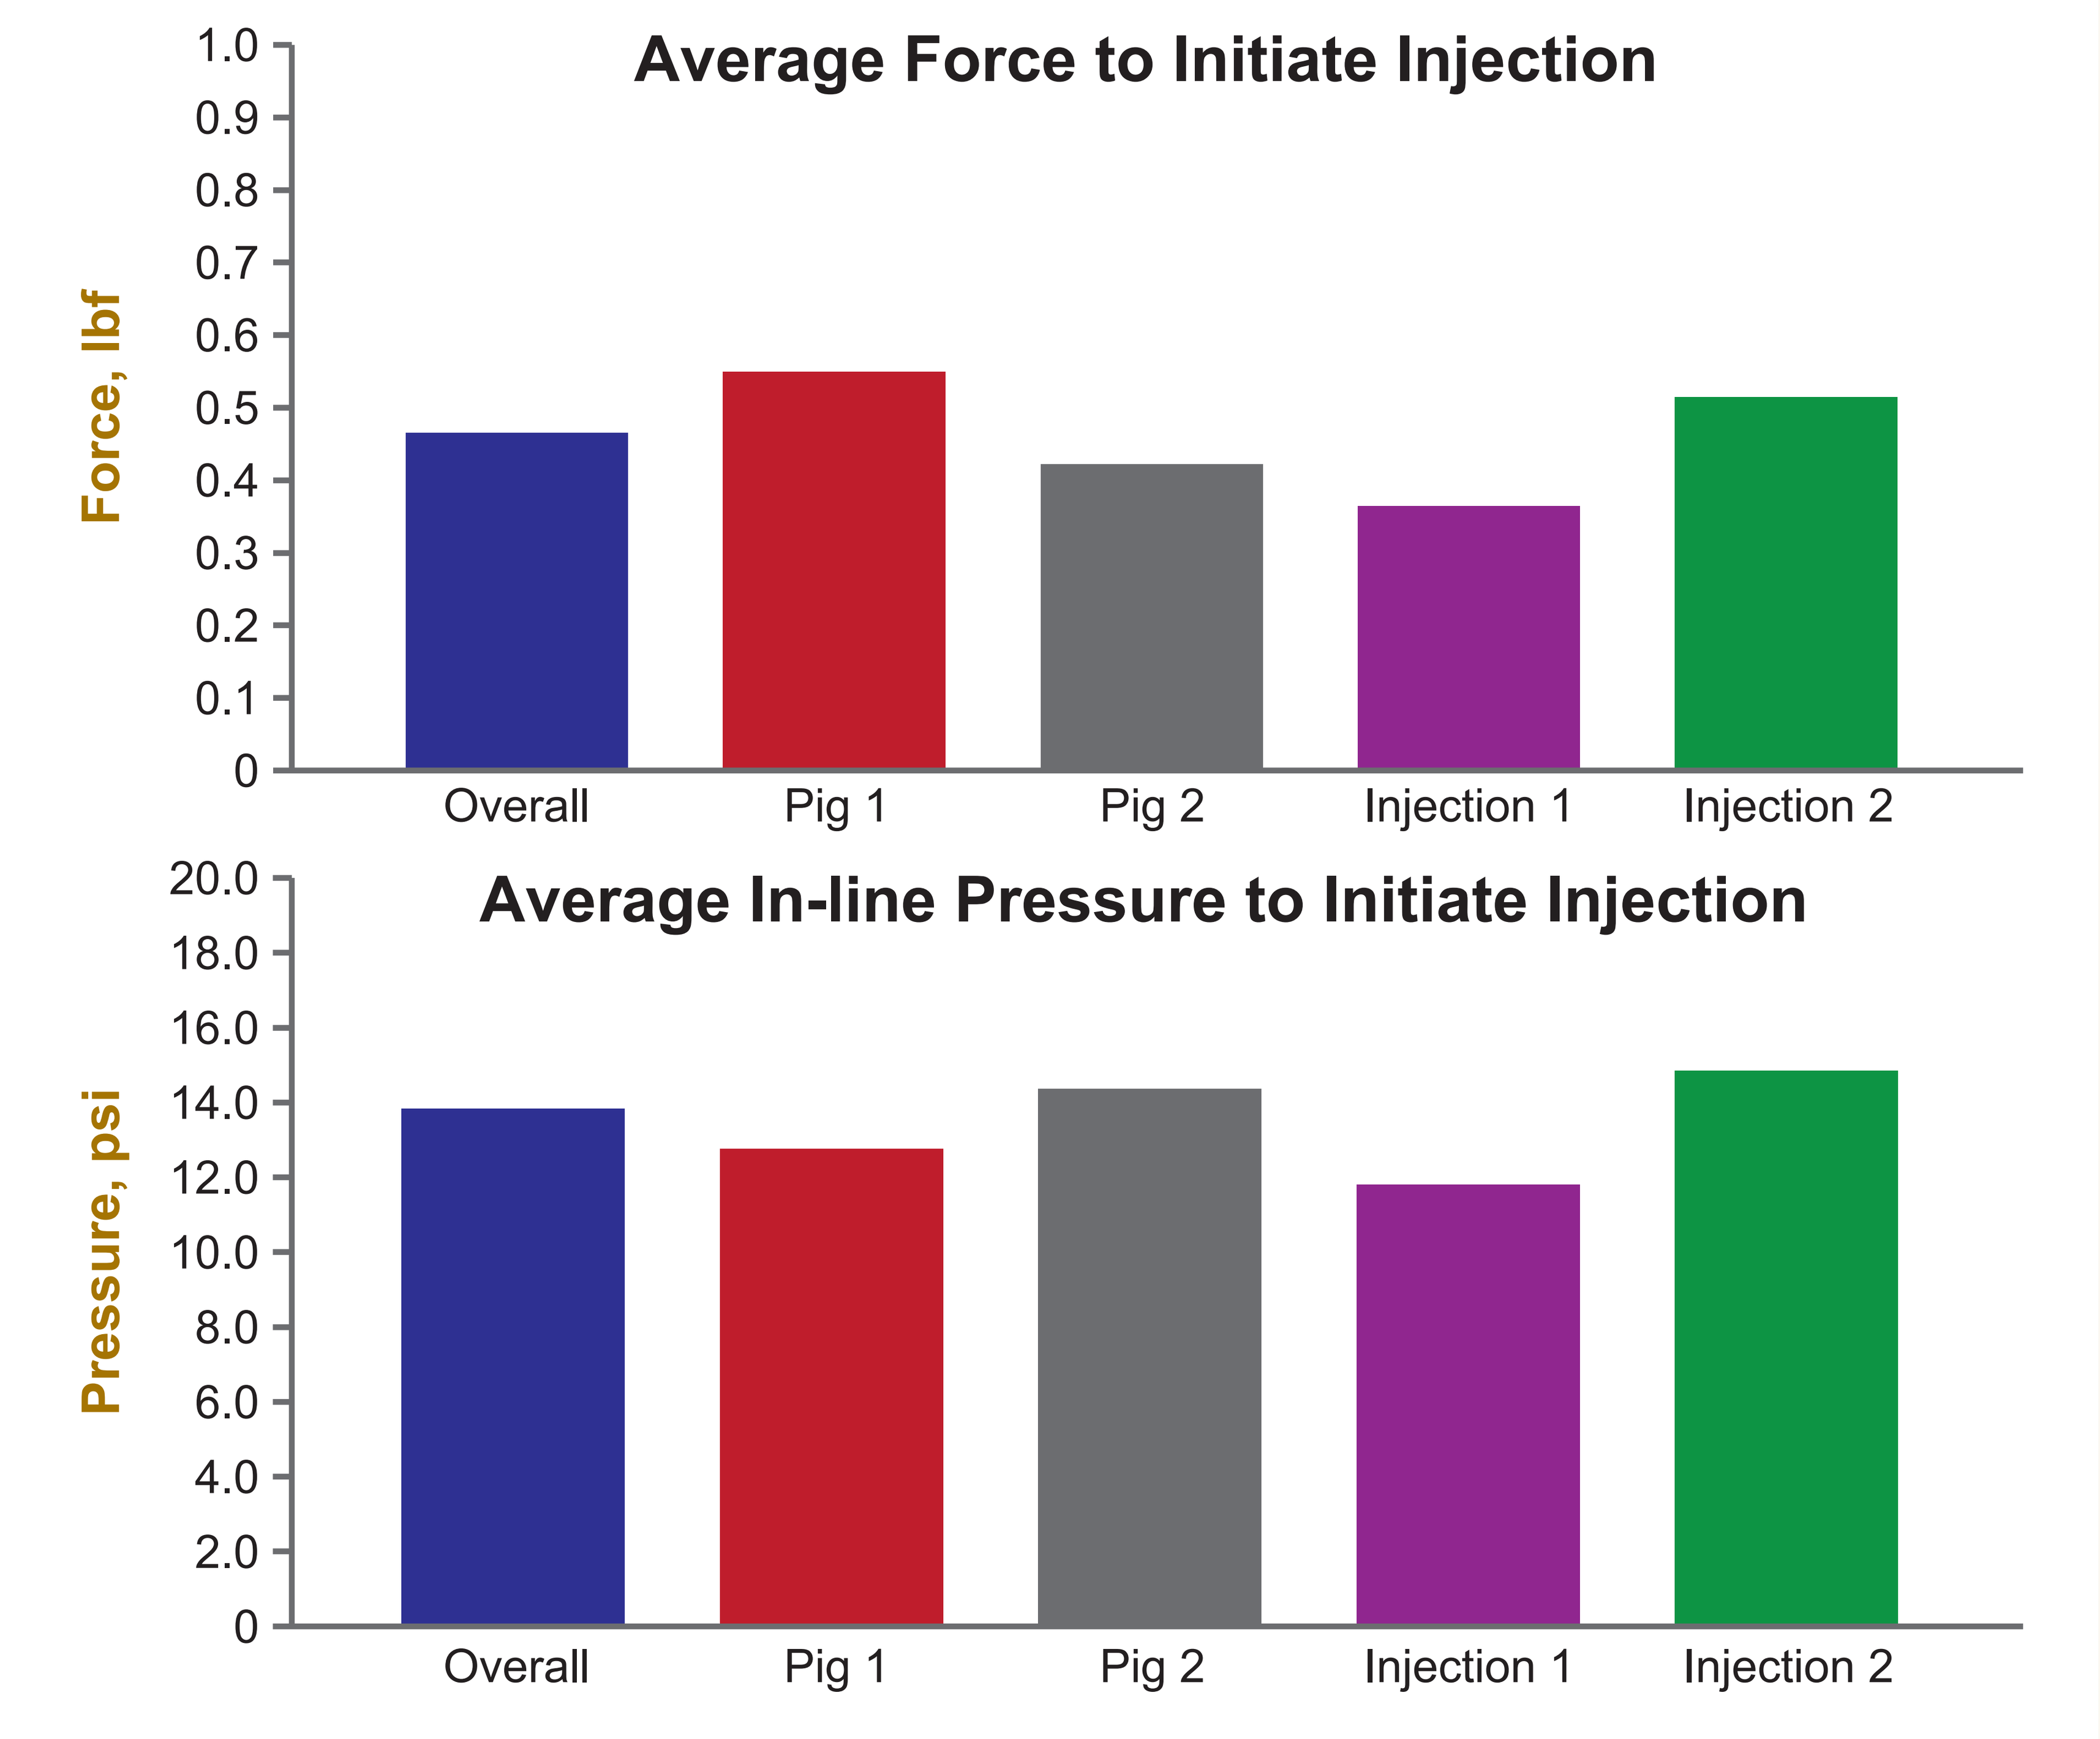
Average Force to Initiate Injection
Force, lbf
Overall
Pig 1
Pig 2
Injection 1
Injection 2
Average In-line Pressure to Initiate Injection
Pressure, psi

Discussion

- This exploratory study found the average overall force required to initiate a suprachoroidal injection—0.465 lbf (4.45 N)—to be well within the physical ability of the user without the assistance of outside mechanical forces (Figure 3). The average overall in-line pressure generated during the initiation of a suprachoroidal injection was observed as 13.83 psi (95.35 kPa). The CFD model provides insight into the fluid dynamics involved in the dissipation of the pressure buildup within the device. The model suggests that the in-line pressure differs greatly from the pressure of the injectate when exposed to the tissues of the eye, specifically at the needle tip. Despite the average pressures within the syringe during an injection procedure reaching as high as 30 times that of normal intraocular pressure, the CFD model demonstrates that pressure of the injectate rapidly drops to ~0.2 psi (1.38 kPa) by the time it exits the needle orifice and enters the suprachoroidal space. The in-line pressure values measured in this study should not be cause for concern, as they do not directly transfer into the ocular tissues during a suprachoroidal injection.
- Three sources of potential variability were identified in the data. Differences in injection technique behaviors between multiple injecting investigators led to different patterns in the injection data when graphed. Additionally, the locations of the instrumentation on the microinjector limited the ways in which the gathered data could be applied. Finally, anatomic variance within the specimens contributed to differences in the data. Two of the 8 original datasets were excluded due to technical difficulties with the equipment setup.

Conclusions

- **Suprachoroidal injections, when performed using a prescribed injection procedure, require no external aid or additional mechanical assistance. Further, this injection procedure is capable of generating and containing forces and pressures adequate to consistently inject fluid into the suprachoroidal space of the eye.**

Disclosures: **N. Fisher, J. Yoo, S. Hancock,** and **R. V. Andino:** Clearside Biomedical Code I (personal financial interest).

Presented at the ARVO 2018 Annual Meeting, April 29–May 3, Honolulu, HI